#148

Rapid and complete allergen delivery by a freeze-dried fast dissolving SLIT-tablet for ragweed allergy immunotherapy

Lawton SA[1], Rance KS[2], Lund K[3], Fonseca H[1], Ohashi-Doi K[4]

[1]ALK A/S, Horsholm, Denmark [2]ALK Americas, Round Rock, US [3]Papermill Medical, Copenhagen, Denmark [4]Torii Pharmaceuticals Co. Ltd., Tokyo, Japan

Background

Allergy immunotherapy (AIT) administered via the sublingual route relies on complete allergen release within the recommended sublingual holding time. According to a European Medicines Agency (EMA) reflection paper[1], orodispersible preparations with rapid dispersion are "*preferred*" for children of 2-5 years and "*the dosage form of choice*" for children of 6-11 years. Rapid and complete allergen release has previously been reported for both the freeze-dried fast dissolving grass and house dust mite (HDM) sublingual immunotherapy (SLIT)-tablet under in-vitro conditions (Figure 1).[2,3] Recently, the freeze-dried fast dissolving ragweed SLIT-tablet was approved for use in children (5-17 years) by the FDA for ragweed-pollen induced allergic rhinitis with or without conjunctivitis, after the efficacy and safety profile was confirmed in the largest clinical AIT study conducted in children to date (n=1025).

Methods

Disintegration time of the ragweed SLIT-tablet (ALK A/S, Denmark) was measured by depositing the tablet into a submersible mesh-basket and agitating at 37°C in an assay buffer (100 mM phosphate, pH 6.8, 0.125% casein). The assay buffer resembles human saliva with regards to pH, ionic strength, and total protein content. Allergen release kinetics in the assay buffer was measured using a mini-vessel (200 mL) Distek Model 2500 instrument (Distek Inc., USA). Amb a 1 was measured by ELISA (Indoor Biotechnologies, USA) at 15, 30, 60, 90, 120, 180 and 300 seconds. Total allergen release was confirmed by comparison to the manufacturer's in-house reference.

1. EMEA/CHMP/PEG/194810/2005
2. Lund K et al. *Clin Ther* 2019;41:742-53
3. Ohashi-Doi K et al. *Int Arch Allergy Immunol* 2017;174:26-34

Figure 1

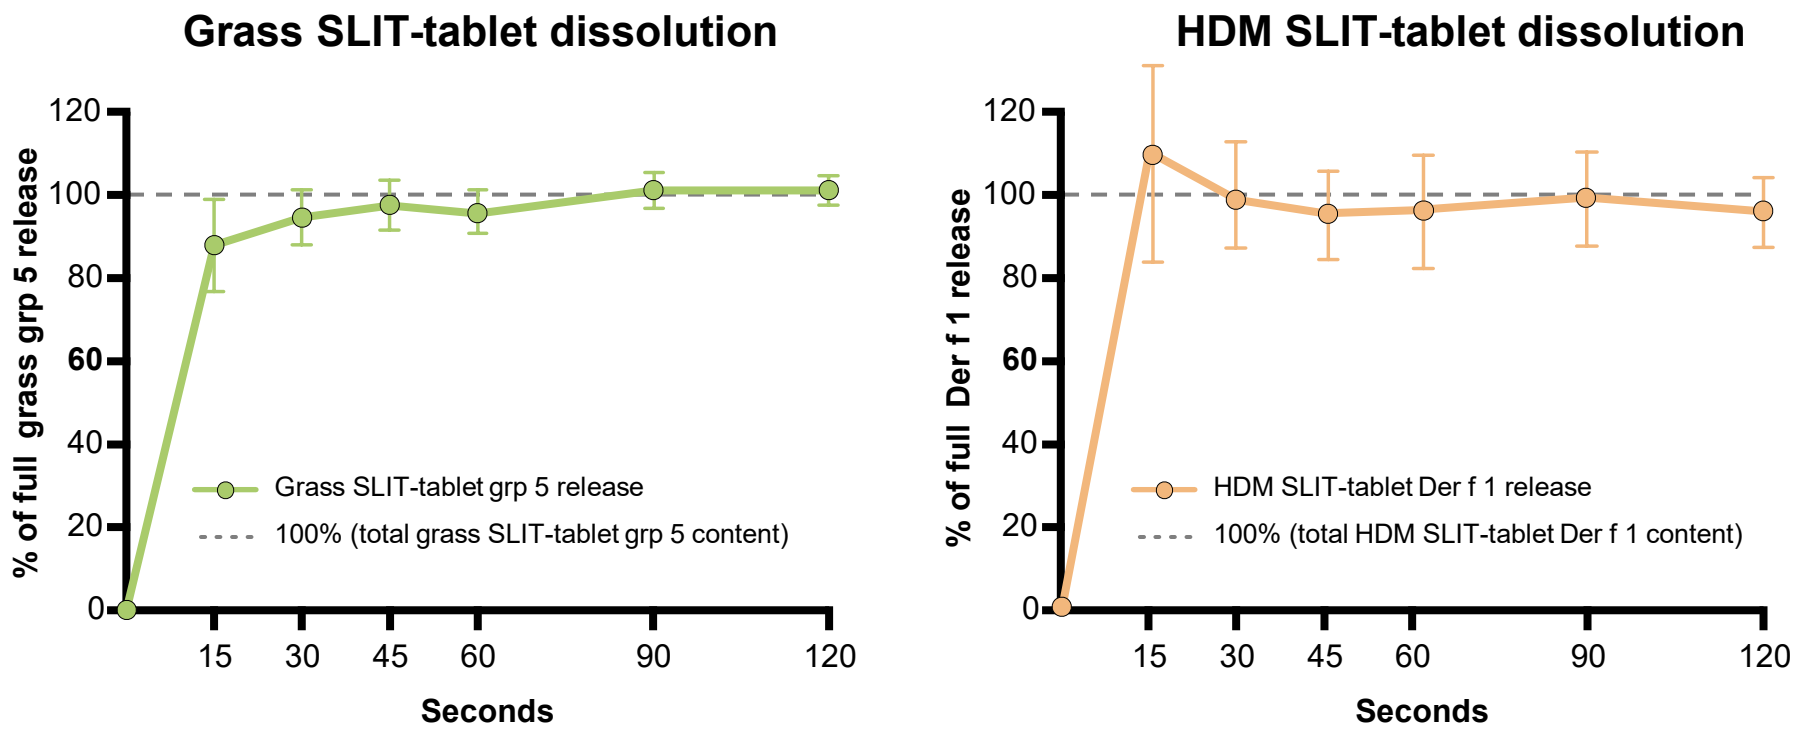
Grass SLIT-tablet dissolution
% of full grass grp 5 release
120
100
80
60
40
20
0
15
30
45
60
90
120
Seconds
Grass SLIT-tablet grp 5 release
100% (total grass SLIT-tablet grp 5 content)
HDM SLIT-tablet dissolution
% of full Der f 1 release
HDM SLIT-tablet Der f 1 release
100% (total HDM SLIT-tablet Der f 1 content)

Figure 2

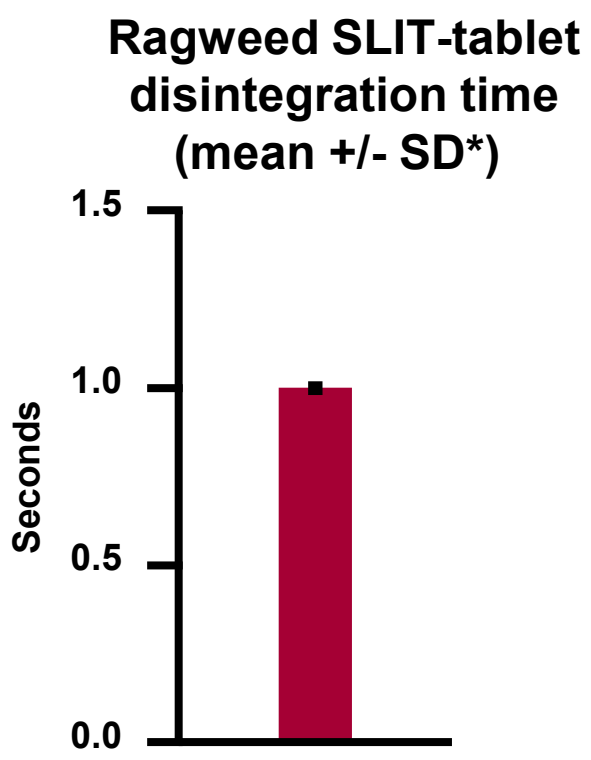
Ragweed SLIT-tablet disintegration time (mean +/- SD*)
1.5
1.0
0.5
0.0
Seconds

*Standard deviation, N=6

Figure 3

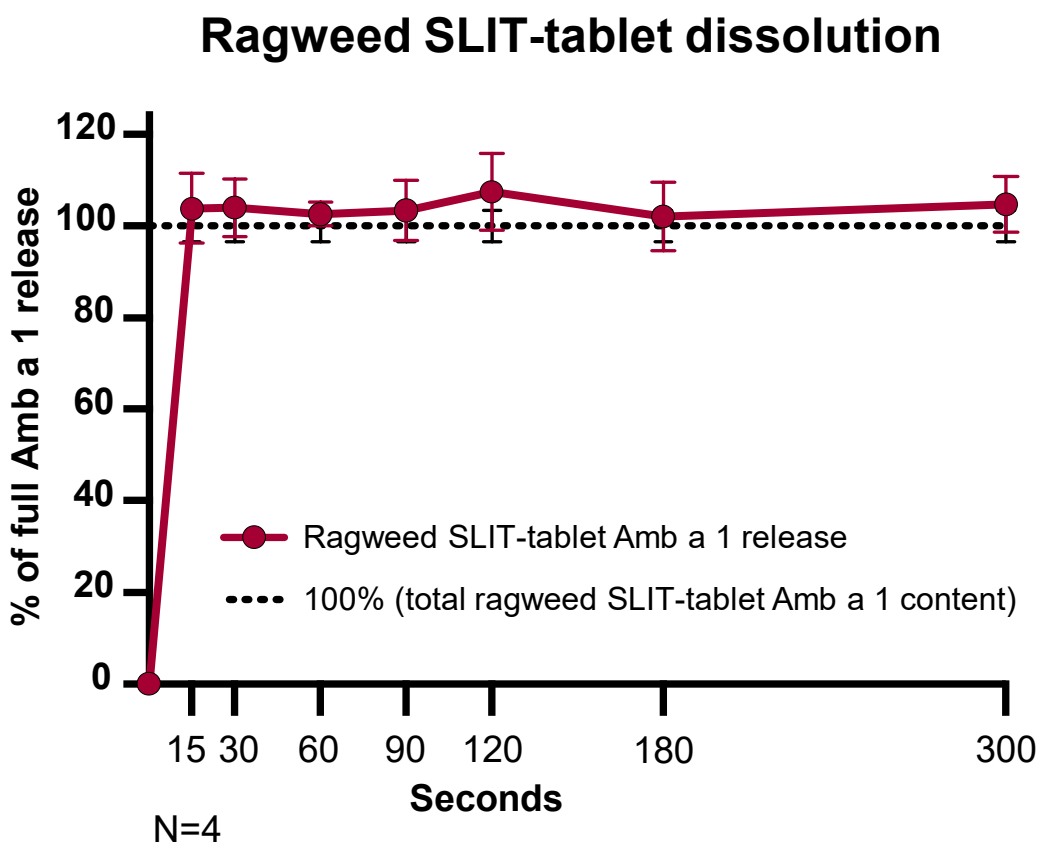
Ragweed SLIT-tablet dissolution
% of full Amb a 1 release
120
100
80
60
40
20
0
15
30
60
90
120
180
300
Seconds
Ragweed SLIT-tablet Amb a 1 release
100% (total ragweed SLIT-tablet Amb a 1 content)

N=4

Results

Complete disintegration of the ragweed SLIT-tablet occurred within 1 second (Figure 2) when deposited into the assay buffer. No tablet residue could be detected.

The ragweed SLIT-tablet achieved complete allergen dissolution and full recovery of soluble Amb a 1 compared to the manufacturer's in-house reference within the first 15 seconds after depositing the tablet into the assay buffer (Figure 3).

Conclusion

The dissolution properties of the freeze-dried fast dissolving ragweed SLIT-tablet ensure full release of the SLIT-tablet allergen content and support short sublingual holding times, which may be of particular importance when treating children

Funding source: ALK A/S, Denmark